Effect and Safety of Patisiran in Patients with Hereditary Transthyretin Amyloidosis with Polyneuropathy and Chronic Kidney Disease

Julien Dang[1], Céline Labeyrie[2], Shaun Bender[3], Alexia Bakdache[4], Marianne T. Sweetser[3], Cécile Cauquil[2]

[1]Nephrology Department, APHP, CHU Bicêtre, Université Paris-Saclay, France; [2]Neurology Department, APHP, CHU Bicêtre, Université Paris-Saclay, France; [3]Alnylam Pharmaceuticals, Cambridge, MA, USA; [4]Alnylam Pharmaceuticals, France.

For US HCPs Only
Scan to View Congress
Material Presented

Conclusions

- **During long-term patisiran treatment, estimated glomerular filtration rate (eGFR) remained relatively stable in patients with transthyretin amyloidosis (ATTR), irrespectively of baseline kidney function and cardiac function**
- **No new safety signals were observed with patisiran treatment in patients with impaired renal function at baseline, including patients with liver transplant**
- **This suggests that patisiran can be a safe and effective treatment option in patients with ATTR and mild-to-moderate concurrent chronic kidney disease (CKD), a common comorbidity in this patient population**
 - **Future studies should focus on longer placebo-controlled trial time to assess the efficacy of gene silencers on renal function, collecting real-world data on efficacy in patients with nephrotic syndrome, and safety for those with eGFR <15 mL/min/1,73 m² (as this could not be assessed in the present study)**

Introduction

- Transthyretin amyloidosis (ATTR) is a progressive, multisystem, and ultimately fatal disease encompassing hereditary ATTR (ATTRv, v for variant) and wild-type ATTR (ATTRwt)[1–4]
 - ATTR is caused by accumulation of misfolded transthyretin (TTR) protein into toxic amyloid fibrils that deposit in multiple organs and tissues, including the heart, peripheral nerves, gastrointestinal tract, and kidneys[1–5]
 - ATTRwt is associated with aging and ATTRv results from genetic variants in the *TTR* gene[1,2]
- Patients with ATTR frequently develop CKD,[6,7] and the proportion of patients with comorbid CKD is likely to increase as patients live longer due to new treatment options
 - However, there are limited data on treatment outcomes in patients with ATTR and concurrent CKD[6,7]
- Patisiran is an intravenously administered RNA interference therapeutic approved for the treatment of ATTRv with polyneuropathy[8,9]

Objective

- To assess the efficacy, on renal function and TTR knockdown, and safety of patisiran in patients with ATTRv or ATTRwt, with and without comorbid CKD

Methods

- This post hoc analysis was performed using data from the following patisiran clinical trials:
 - The Phase 3 APOLLO study (NCT01960348), a randomized, double-blind, placebo-controlled trial in patients with ATTRv with polyneuropathy[9]
 - The Phase 3 APOLLO-B study (NCT03997383), a randomized, double-blind, placebo-controlled trial in patients with ATTRwt and ATTRv with cardiomyopathy[10]
 - A Phase 3b open-label study (NCT03862807) in patients with ATTRv with polyneuropathy who had disease progression after undergoing orthotopic liver transplant (OLT)[11]
 - A Phase 2 open-label extension (OLE) study (NCT01961921) in patients with ATTRv with polyneuropathy[12]
 - A global OLE study (NCT02510261) in patients with ATTRv with polyneuropathy who had completed the APOLLO or Phase 2 OLE studies[13]
- The analysis assessed:
 - eGFR and TTR levels in patients with baseline eGFR <60 vs ≥60 mL/min/1.73 m²
 - eGFR by baseline cardiac function (longitudinal strain <–15% vs ≥–15%)
 - Safety in patients with baseline eGFR <60 mL/min/1.73 m²

Results

Patients

- A total of 634 patients with ATTR were included in the analysis, 225 from APOLLO, 359 from APOLLO-B, 27 from the Phase 2 OLE, and 23 from the post-OLT study (**Table 1**)
 - Of these, 346 (54.6%) had ATTRv and 288 (45.4%) had ATTRwt
 - Patients with ATTRwt tended to be older than those with ATTRv
- Across the studies, 158 (24.9%) patients had eGFR <60 mL/min/1.73 m² at baseline, with a higher incidence observed in the APOLLO-B and post-OLT study compared with APOLLO and the Phase 2 OLE (**Table 1**)
 - This may reflect the older age and worse cardiac function in APOLLO-B, and the higher use of concomitant immunosuppressants in the post-OLT study, compared with the other studies
- Baseline longitudinal strain ≥–15% was observed in 404 (63.7%) patients, and was more common in the APOLLO-B study compared with the other studies

Table 1. Baseline Demographics and Disease Characteristics

Characteristic	ATTRv with polyneuropathy				ATTR with cardiomyopathy			
	Phase 2 OLE	APOLLO		Post-OLT study	APOLLO-B			
					ATTRv		ATTRwt	
	Patisiran (n=27)	Placebo (n=77)	Patisiran (n=148)	Patisiran (n=23)	Placebo (n=34)	Patisiran (n=37)	Placebo (n=144)	Patisiran (n=144)
Median age at screening (range), year	64 (29–77)	63 (34–80)	62 (24–83)	58 (43–75)	66 (41–85)	70 (47–85)	77 (59–85)	77 (59–85)
NYHA Class, n (%)								
No heart failure[a]	N/A	N/A	N/A	13 (65.5)	N/A	N/A	N/A	N/A
I	19 (70.4)	40 (52.0)	70 (47.0)	5 (21.7)	4 (11.8)	2 (5.4)	11 (7.6)	8 (5.6)
II	7 (25.9)	36 (47.0)	77 (52.0)	5 (21.7)	28 (82.4)	33 (89.2)	122 (84.7)	123 (85.4)
III	0	0	0	0	2 (5.9)	2 (5.4)	11 (7.6)	13 (9.0)
Missing	1 (3.7)	1 (1.0)	1 (<1.0)	0	0	0	0	0
eGFR at baseline, n (%) (min-max)								
<60 mL/min/1.73 m²	1 (3.7) (58–58)	5 (6.5) (32–60)	16 (10.8) (31–60)	9 (39.1) (37–56)	8 (23.5) (23–59)	10 (27.0) (31–59)	61 (42.4) (27–59)	48 (33.3) (30–59)
≥60 mL/min/1.73 m²	26 (96.3) (62–153)	72 (93.5) (60–228)	132 (89.2) (60–346)	14 (60.9) (63–114)	26 (76.5) (61–162)	24 (64.9) (61–223)	81 (56.3) (60–162)	95 (66.0) (60–138)
***TTR* genotype, n (%)**								
V30M	20 (74.1)	40 (51.9)	56 (37.8)	15 (65.2)	1 (2.9)	1 (2.7)	N/A	N/A
Non-V30M	7 (25.9)	37 (48.1)	92 (62.2)	8 (34.8)	33 (97.1)	36 (97.3)	N/A	N/A
Longitudinal strain at baseline, n (%)[b]								
<–15%	21 (77.8)	46 (59.7)	87 (58.8)	5 (21.7)	5 (14.7)	5 (13.5)	12 (8.3)	14 (9.7)
≥–15%	6 (22.2)	26 (33.8)	51 (34.5)	5 (21.7)	29 (85.3)	32 (86.5)	127 (88.2)	128 (88.9)

[a]For the Phase 2 OLE and APOLLO studies, NYHA Class I included patients with no heart failure and patients with heart failure who had no symptomatology during ordinary physical activity; [b]Percentages may not add to 100% where baseline longitudinal strain data were not available for all patients.

Efficacy Outcomes

- During long-term patisiran treatment, mean eGFR remained relatively stable irrespective of baseline eGFR (**Figure 1A–C**)
- eGFR levels also remained stable regardless of baseline longitudinal strain (**Figure 2A–B**)
- Patisiran resulted in rapid knockdown of serum TTR that was consistent, irrespective of baseline eGFR, and the knockdown was maintained throughout the studies (**Figure 3A–B**)

Safety Outcomes

- Patisiran had an acceptable safety profile in patients with baseline eGFR <60 mL/min/1.73 m² across the studies, with no new safety findings observed (**Table 2** and **Table 3**)

Figure 1. Change Over Time in Mean eGFR by Baseline eGFR Subgroups

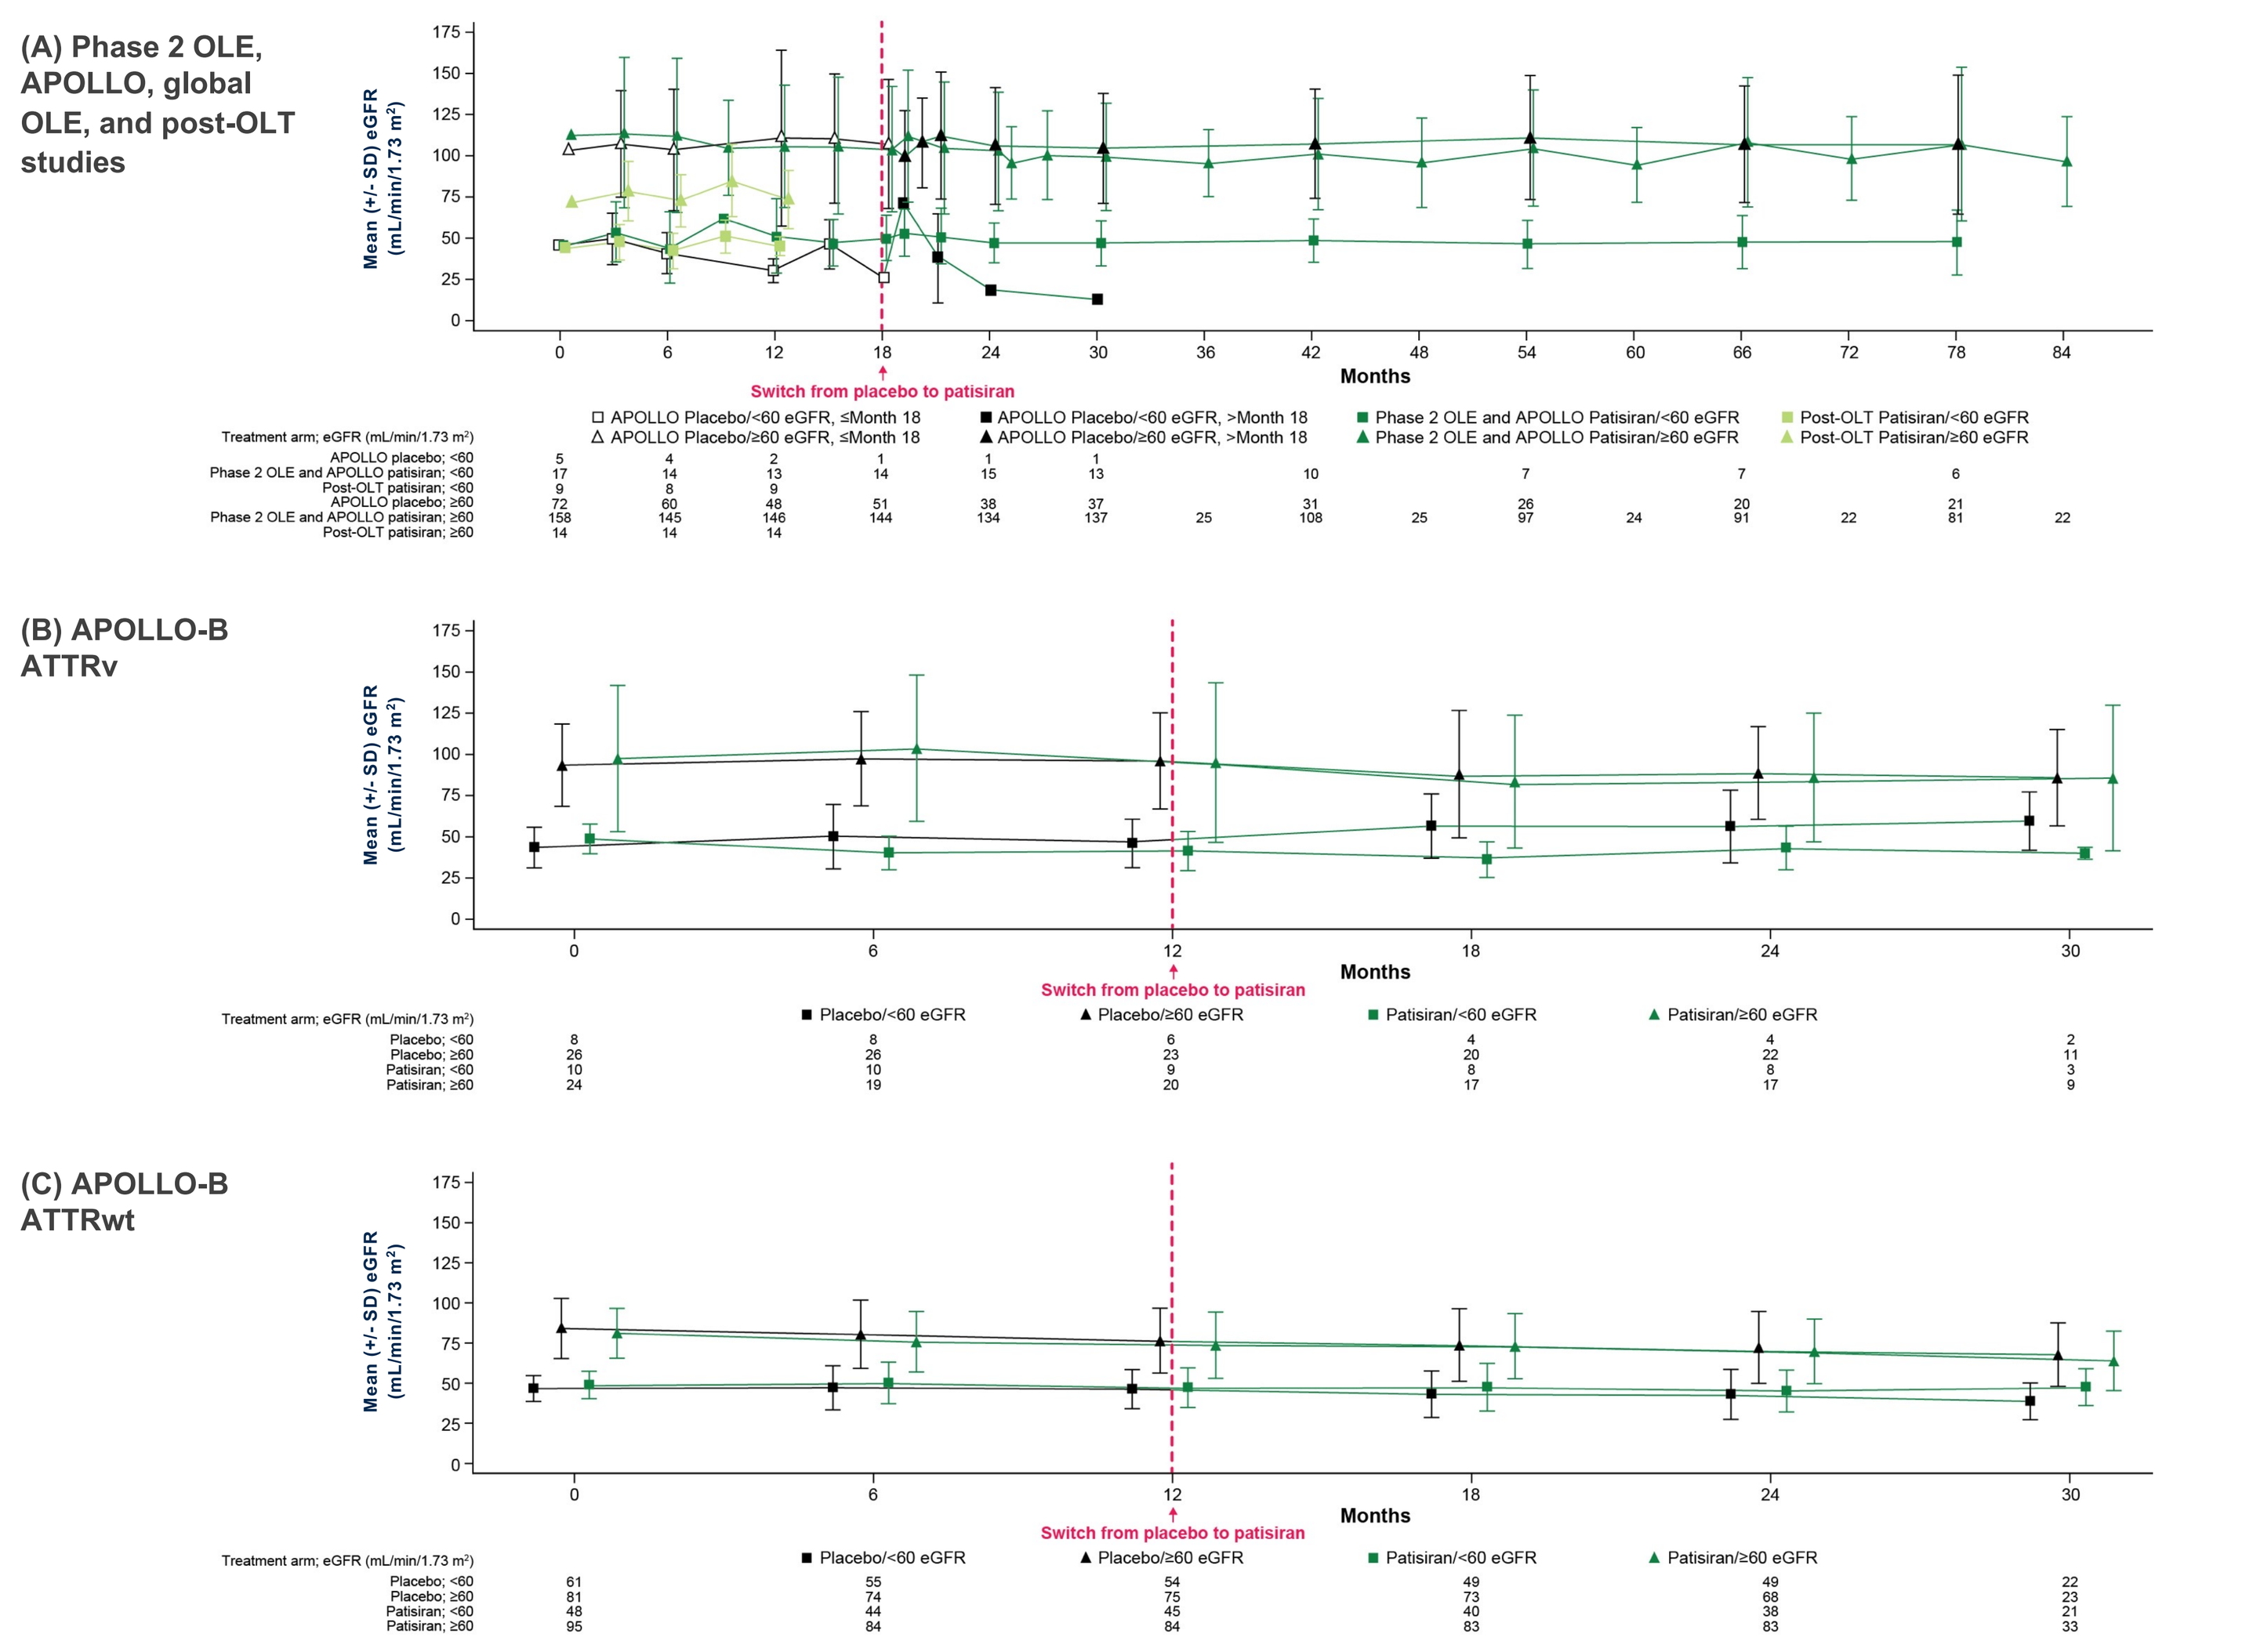
(A) Phase 2 OLE, APOLLO, global OLE, and post-OLT studies
(B) APOLLO-B ATTRv
(C) APOLLO-B ATTRwt
Mean (+/- SD) eGFR (mL/min/1.73 m²)
Months
Switch from placebo to patisiran

Figure 2. Change Over Time in Mean eGFR by Baseline Longitudinal Strain Subgroups

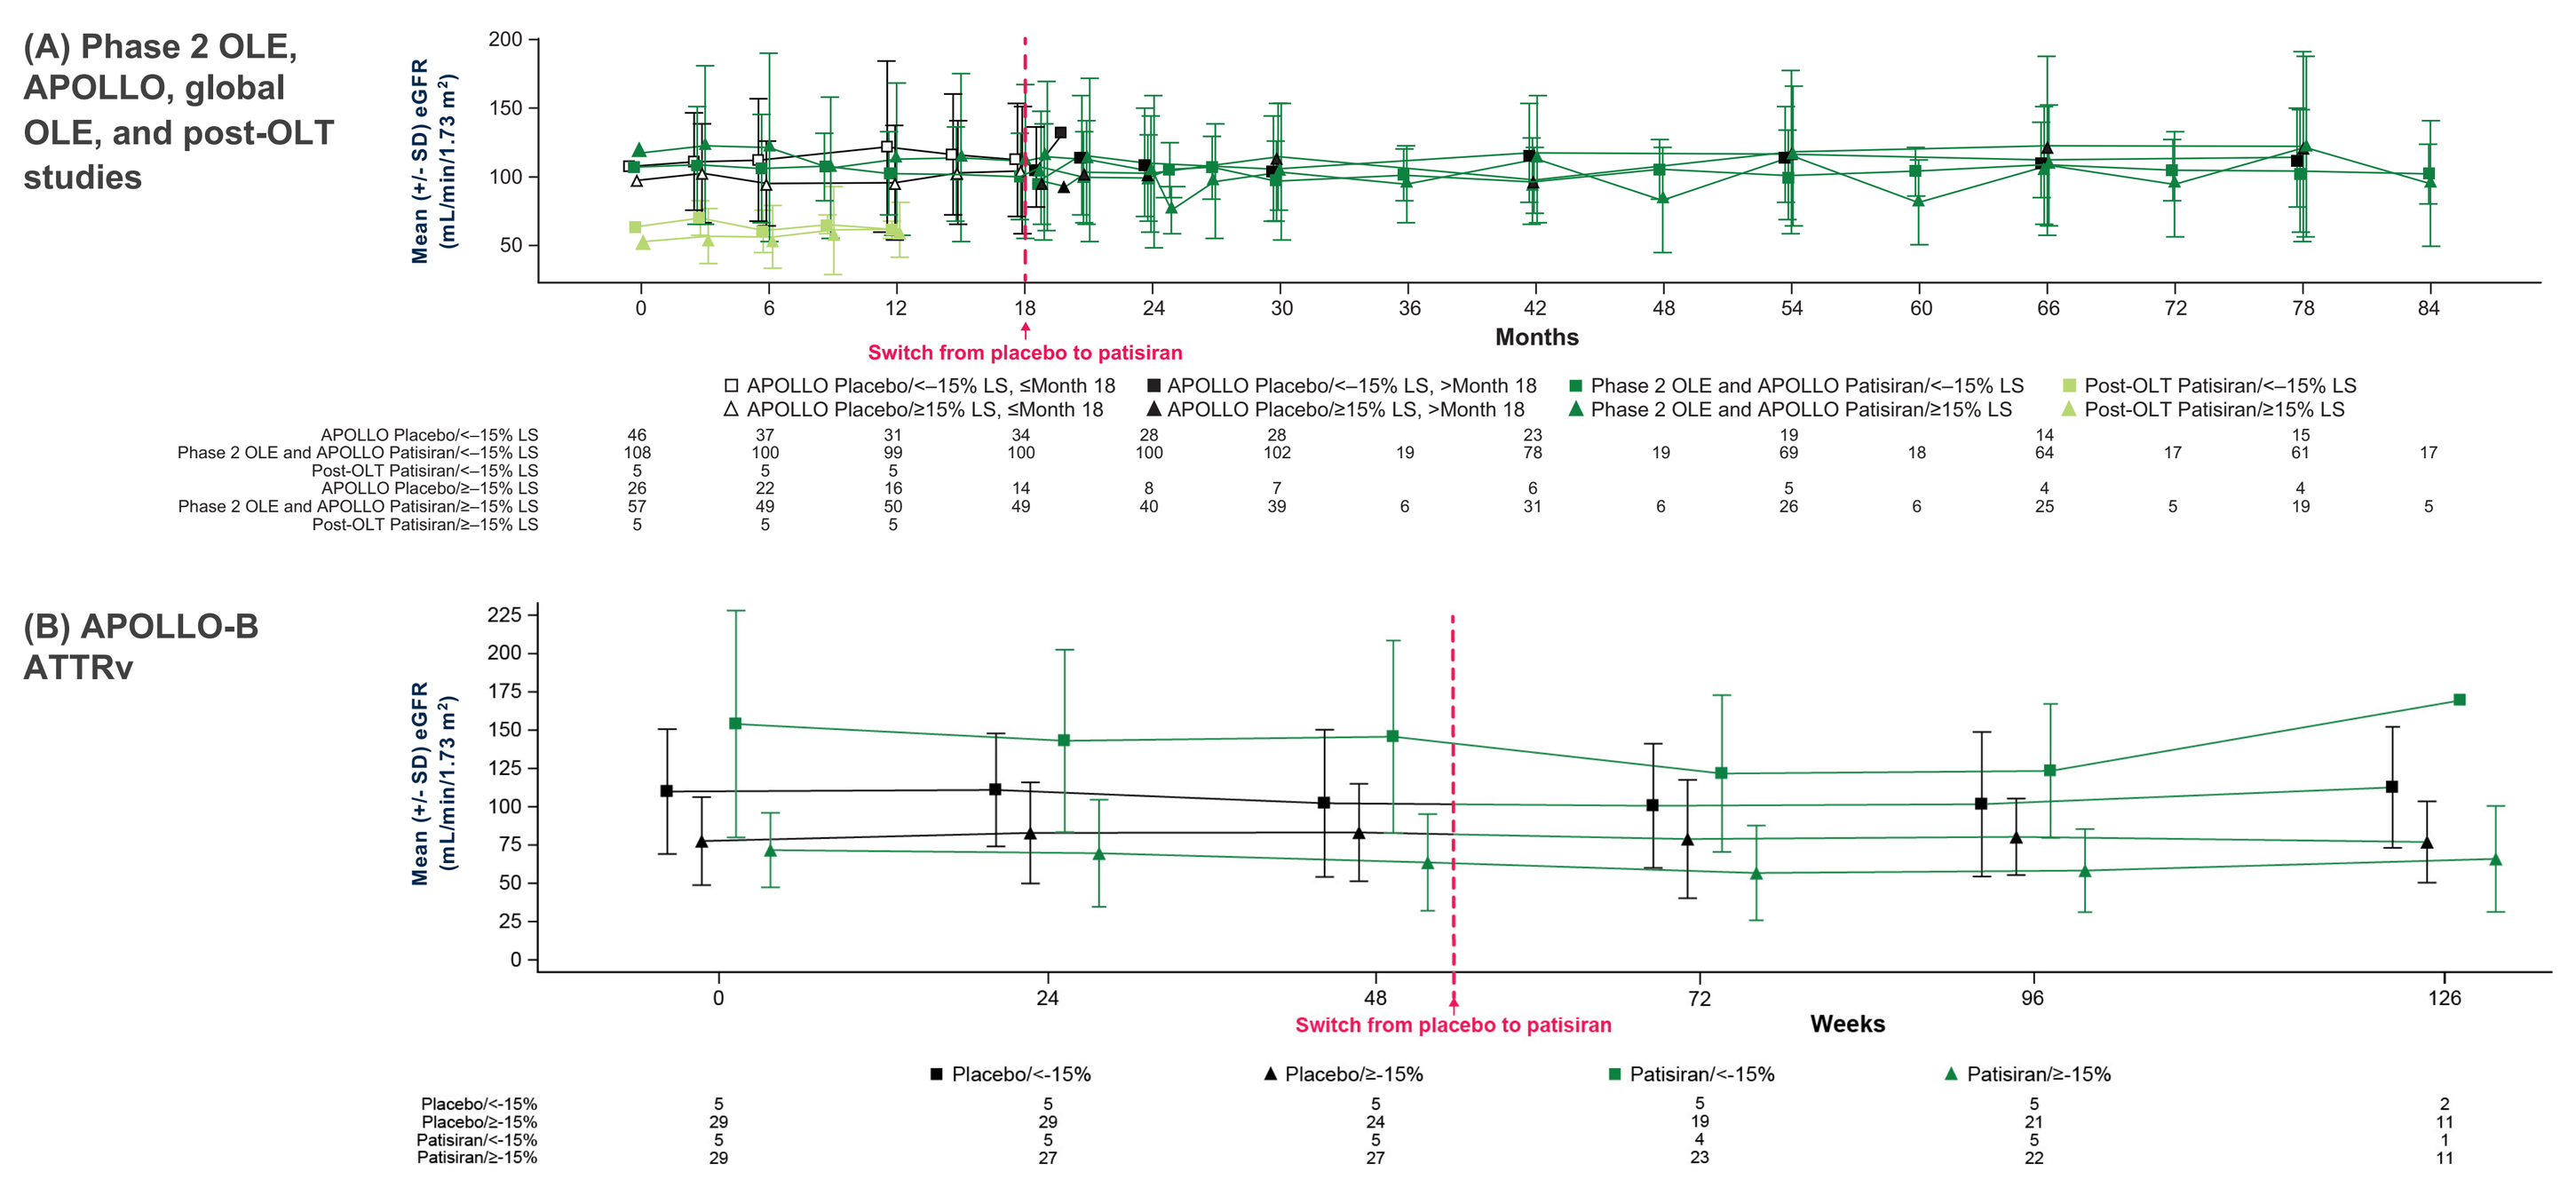
(A) Phase 2 OLE, APOLLO, global OLE, and post-OLT studies
(B) APOLLO-B ATTRv
Mean (+/- SD) eGFR (mL/min/1.73 m²)
Months
Weeks
Switch from placebo to patisiran

Figure 3. Serum TTR Knockdown by Baseline eGFR

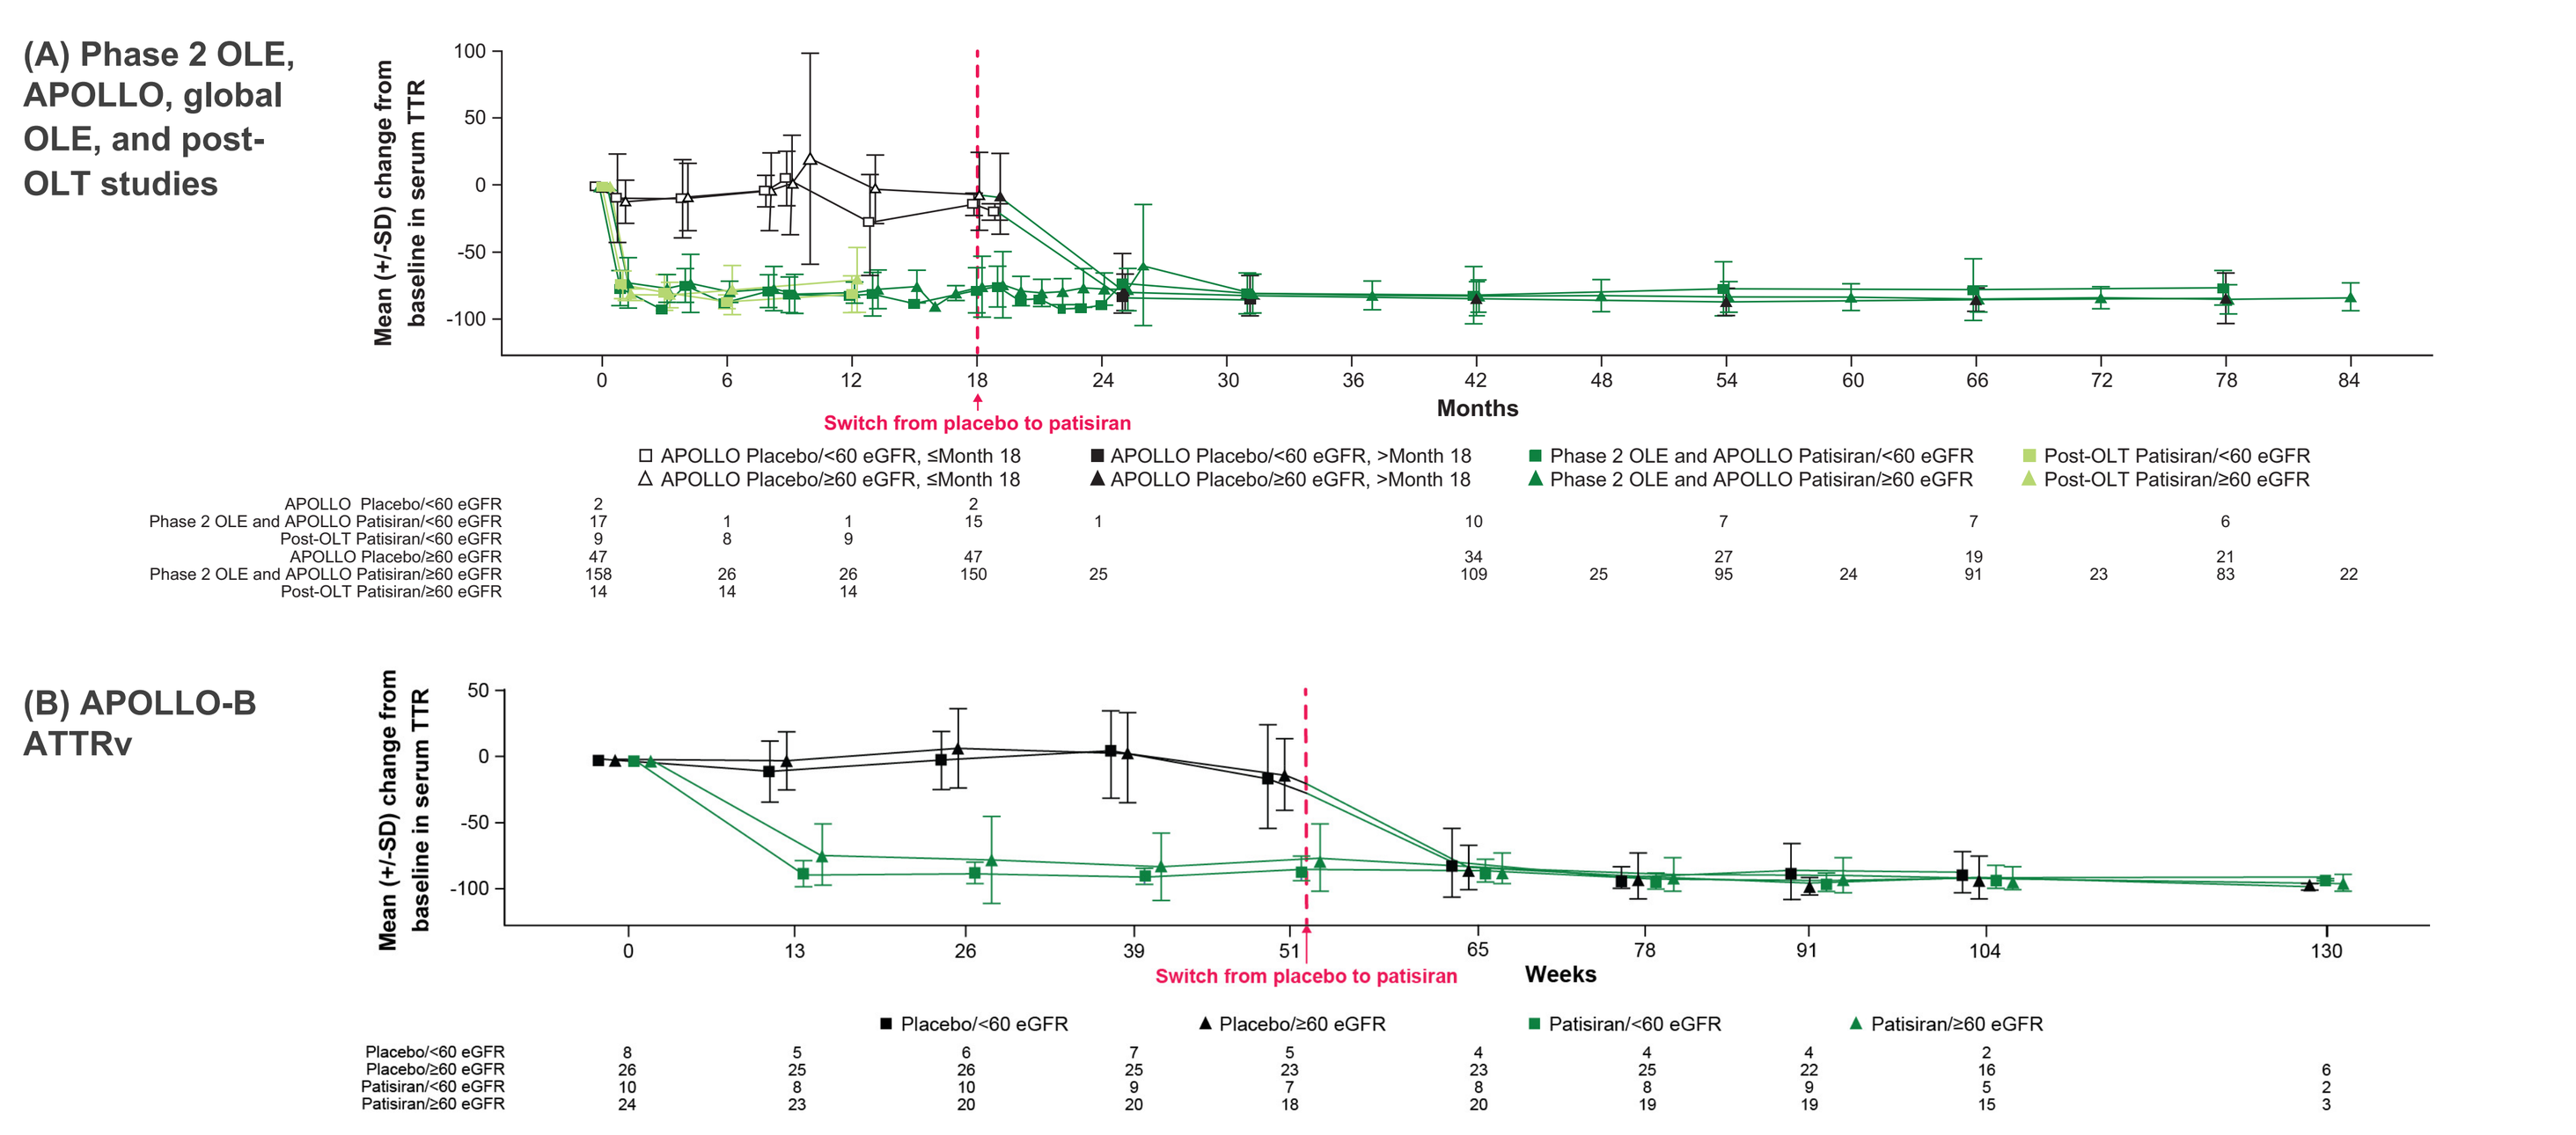
(A) Phase 2 OLE, APOLLO, global OLE, and post-OLT studies
(B) APOLLO-B ATTRv
Mean (+/-SD) change from baseline in serum TTR
Months
Weeks
Switch from placebo to patisiran

Table 2. Renal and Urinary AEs During Treatment with Patisiran in Patients with ATTRv with Polyneuropathy and Baseline eGFR <60 mL/min/1.73 m²

Event	ATTRv with polyneuropathy			
	Phase 2 OLE/ Global OLE	APOLLO/Global OLE		Post-OLT study
	Patisiran (n=1) [PY=2.1]	Placebo/patisiran (n=5)[a] [PY=6.8]	Patisiran (n=16) [PY=73.2]	Patisiran (n=9) [PY=9.8]
≥1 AE, n (%)	0	2 (40.0)	7 (43.8)	0
Renal and urinary AEs, n (%)	0	2 (40.0)	7 (43.8)	0
Acute kidney injury	0	0	2 (12.5)	0
Chronic kidney disease	0	0	2 (12.5)	0
Dysuria	0	1 (20.0)	2 (12.5)	0
End stage renal disease	0	1 (20.0)	0	0
Hematuria	0	1 (20.0)	1 (6.3)	0
Neurogenic bladder	0	0	1 (6.3)	0
Oliguria	0	0	1 (6.3)	0
Renal failure	0	0	0	0
Renal impairment	0	1 (20.0)	0	0
Urinary incontinence	0	0	1 (6.3)	0
Urinary retention	0	1 (20.0)	0	0

[a]Placebo patients switched to patisiran at 18 months after entering the global OLE. Only events occurring on patisiran are counted.

Table 3. Renal and Urinary AEs During the 12-Month Double-Blind Period in Patients with ATTRv with Cardiomyopathy and Baseline eGFR <60 mL/min/1.73 m²

Event	ATTRv with cardiomyopathy	
	APOLLO-B ATTRv	
	Placebo (n=8) [PY=7.6]	Patisiran (n=10) [PY=10.3]
≥1 AE, n (%)	8 (100.0)	9 (90.0)
Renal and urinary AEs, n (%)	2 (25.0)	3 (30.0)
Acute kidney injury	0	0
Chronic kidney disease	0	0
Dysuria	0	0
End stage renal disease	0	0
Hematuria	0	1 (10.0)
Neurogenic bladder	0	0
Oliguria	0	0
Renal failure	1 (12.5)	0
Renal impairment	1 (12.5)	1 (10.0)
Urinary incontinence	0	0
Urinary retention	0	1 (10.0)

Thank you to the patients, their families, Investigators, study staff, and collaborators for their participation.
Acknowledgments: Medical writing assistance was provided by John Ewbank, PhD, on behalf of Adelphi Communications Ltd, UK, and funded by Alnylam Pharmaceuticals in accordance with Good Publication Practice guidelines.

Disclaimer: Patisiran is not approved for the treatment of ATTR-CM in the US, but ANSM has granted approval for the compassionate use of patisiran in France for patients with ATTR-CM failing tafamidis 61 mg.

References: 1. Hawkins et al. *Ann Med* 2015;47:625–38; 2. Ruberg et al. *J Am Coll Cardiol* 2019;73:2872–91; 3. Maurer et al. *J Am Coll Cardiol* 2016;68:161–72; 4. Adams et al. *Nat Rev Neurol* 2019;15:387–404; 5. Dang et al. *Nephrol Dial Transplant* 2023;38:2019–2030; 6. Solignac J et al. *Clin Kidney J* 2022;15:1747–54; 7. Dang J et al. *Amyloid* 2023;30:38–48; 8. Kristen et al. *Neurodegener Dis Manag* 2019;9:5–23; 9. Adams et al. *N Engl J Med* 2018;379:11–21; 10. Maurer et al. *N Engl J Med* 2023;389:1553–65; 11. Schmidt et al. *Am J Transplant* 2022;22:1646–57; 12. Coelho et al. *Orphanet J Rare Dis* 2020;15:179; 13. Adams et al. *Lancet Neurol* 2021;20:49–59.

Presented at The Annual Meeting of the International Society of Amyloidosis (ISA) 2024, Rochester, MN, USA; May 26–30, 2024.

Abbreviations: AE, adverse event; ATTRv, hereditary transthyretin-mediated, v for variant; ATTRwt, wild-type transthyretin amyloidosis; CKD, chronic kidney disease; eGFR, estimated glomerular filtration rate; N/A, not applicable; NYHA, New York Heart Association; OLE, open label extension; OLT, orthotic liver transplant; SD, standard deviation; TTR, transthyretin

Funding: This study was funded by Alnylam Pharmaceuticals.

Disclosures: **C.L.** and **C.C.** report honoraria from Alnylam Pharmaceuticals. **S.B.**, **A.B.**, and **M.T.S.** are employees of Alnylam Pharmaceuticals and own stock and stock options. **J.D.** reports no conflicts of interest.